395

First-in-human Phase 1 dose-escalation study of AV-203, a monoclonal antibody against ERBB3 in patients with metastatic or advanced solid tumors

John Sarantopoulos,[1] Michael Gordon,[2] R. Donald Harvey,[3] Kamalesh Sankhala,[1] Laeeq Malik,[1] Devalingam Mahalingam,[1] Taofeek K. Owonikoko,[3] Colleen Lewis,[3] Francis Payumo,[4] James Miller,[4] Christine Powell,[4] Zhigang Weng,[4] Philip Komarnitsky,[4] Suresh Ramalingam[3]

[1]Institute for Drug Development, Cancer Therapy and Research Center, University of Texas Health Science Center at San Antonio, San Antonio, TX; [2]Pinnacle Oncology Hematology, Scottsdale, AZ; [3]Winship Cancer Institute of Emory University, Atlanta, GA; [4]AVEO Oncology, Cambridge, MA

Background

- ERBB3, a unique member of the epithelial growth factor receptor (EGFR) tyrosine kinase family, is
 - Aberrantly expressed or activated in multiple human tumors[1,2]
 - Involved in resistance to tyrosine kinase inhibitors[3]
 - A promising target for cancer therapeutics
- AV-203 is a superhumanized IgG1k monoclonal antibody against the ERBB3 receptor (**Figure 1**) with[4–6]
 - High-affinity binding to human and cynomolgus ERBB3[4]
 - No binding to EGFR/ERBB1, ERBB2/HER2, ERBB4/HER4, or murine ERBB3[4]
 - Potent inhibition of both ligand-dependent and -independent ERBB3 signaling[4,5]
 - Dose-dependent inhibition of tumor growth in human cancer models[4,5]
 - Demonstrated ability to restore sensitivity to receptor tyrosine kinase inhibitors[6]

Figure 1. AV-203 Mechanism of Action. NRG-1 binding of ERBB3 causes (A) receptor dimerization and activation, which is (B) blocked by AV-203.

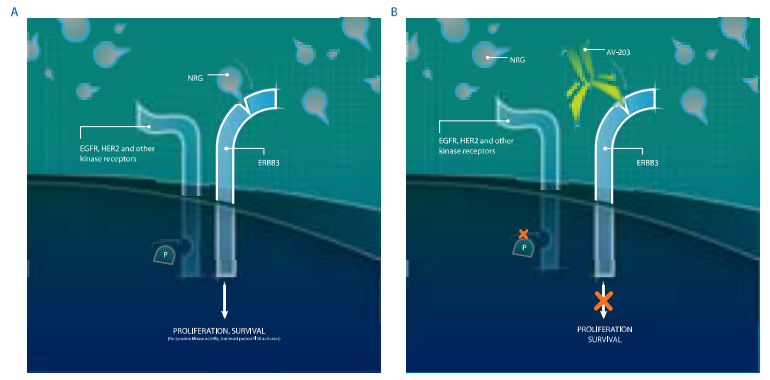
A
B
NRG
AV-203
EGFR, HER2 and other kinase receptors
ERBB3
P
PROLIFERATION, SURVIVAL
PROLIFERATION
SURVIVAL

EGFR, epidermal growth factor receptor 2; HER2, human epidermal growth factor receptor 2; NRG, neuregulin; P13K, phosphatidylinositol 3-kinase.

Study Objective

- The objective of this first-in-human Phase 1 study was to evaluate the safety, tolerability, dose limiting toxicities (DLTs), maximum tolerated dose, and/or recommended Phase 2 dose (RP2D) of AV-203 in patients with advanced solid tumors (NCT01603979)

Methods

- This was a Phase 1 study of AV-203 using a 3 + 3 dose-escalation design
- AV-203 was administered at 2, 5, 9, 14, or 20 mg/kg intravenously once every 2 weeks (1 cycle=28 days, 2 doses per cycle)
- Neuregulin 1 (NRG-1, also known as "heregulin") status was analyzed via RT-PCR analysis of formalin-fixed paraffin-embedded (FFPE) tumor tissue samples (©AltheaDx, Inc., San Diego, CA)
- Pharmacokinetic (PK), anti-drug antibody (ADA), and pharmacodynamic (PD) bioanalyses were performed (PPD®, Richmond, VA)
- Select eligibility requirements include
 - Patients ≥18 years of age who had histologically and/or cytologically confirmed primary diagnosis of advanced solid tumors and had progressed on standard therapies or had no proven treatment options
 - Patients with hypersensitivity to recombinant proteins, significant cardiovascular disease or conduction disturbance, history of severe arrhythmia or familial arrhythmia were excluded
 - Other exclusions included current or prior central nervous system or leptomeningeal metastases, thromboembolic or vascular disorders within 3 months, or positive diagnosis of hepatitis B or C or human immunodeficiency virus
- The primary objective was to determine the RP2D of AV-203; safety was evaluated through the investigation of adverse events (AEs) and DLTs
- Secondary objectives included the investigation of PK and ADA characteristics, and tumor response data
- Exploratory objectives included NRG-1 as a predictive biomarker and PD characteristics

Results

Patients

- A total of 22 patients received AV-203 at doses from 2 mg/kg to 20 mg/kg (**Table 1**)
 - Mean patient age was 63.9 years
 - Eastern Cooperative Oncology Group performance score was 0 or 1
 - A total of 31.8% of patients had received ≤2 prior therapies
- Patients received a mean of 3.5 cycles of AV-203 (**Table 2**)
 - Median time on treatment for all patients was 43 days (range: 1–603)
 - Treatment discontinuations included 16 patients due to progressive disease
- One death occurred on study and was unrelated to AV-203 administration (progressive disease in the 2-mg/kg cohort)

Table 1. Baseline Patient Characteristics

	2 mg/kg (n=7)	5 mg/kg (n=3)	9 mg/kg (n=3)	14 mg/kg (n=3)	20 mg/kg (n=6)	Total (N=22)
Sex, n (%)						
Male	4 (57.1)	2 (66.7)	3 (100.0)	2 (66.7)	4 (66.7)	15 (68.2)
Age						
Mean (SD)	62.3 (12.6)	62.3 (17.4)	64.3 (8.1)	54.3 (20.3)	71.0 (6.4)	63.9 (12.6)
Race, n (%)						
White	6 (85.7)	3 (100.0)	2 (66.7)	3 (100.0)	5 (83.3)	19 (86.4)
Black	0 (0)	0 (0)	1 (33.3)	0 (0)	1 (16.7)	2 (9.1)
ECOG performance score, n (%)						
0	5 (71.4)	1 (33.3)	1 (33.3)	1 (33.3)	0 (0)	8 (36.4)
1	2 (28.6)	2 (66.7)	2 (66.7)	2 (66.7)	6 (100.0)	14 (63.6)
Tumor type, n (%)						
Colorectal	3 (42.9)	0 (0)	1 (33.3)	0 (0)	0 (0)	4 (18.2)
sqNSCLC	0 (0)	1 (33.3)	1 (33.3)	1 (33.3)	1 (16.7)	4 (18.2)
Squamous cell, skin	0 (0)	1 (33.3)	0 (0)	1 (33.3)	0 (0)	2 (9.1)
Other solid tumors[a]	4 (57.1)	1 (33.3)	1 (33.3)	1 (33.3)	5 (83.3)	12 (54.5)
No. of prior therapy,[b] n (%)						
≤2	0 (0)	2 (66.7)	2 (66.7)	1 (33.3)	2 (33.3)	7 (31.8)
3–5	7 (100.0)	0 (0)	1 (33.3)	2 (66.7)	1 (16.7)	11 (50.0)
6–8	0 (0)	0 (0)	0 (0)	0 (0)	2 (33.3)	2 (9.1)
Unknown	0 (0)	1 (33.3)	0 (0)	0 (0)	1 (16.7)	2 (9.1)

ECOG, Eastern Cooperative Oncology Group; SD, standard deviation; sqNSCLC, squamous non-small cell lung cancer.
[a]One each of bladder, cervical, mesothelioma, neuroendocrine tumor of the stomach, non-bone osteosarcoma, stromal tumor-endometrial, pancreatic, ovarian, hepatocellular carcinoma, peritoneal, small cell lung carcinoma, squamous cell carcinoma of the head and neck.
[b]Does not include adjuvant/neoadjuvant therapy.

Table 2. Patient Disposition

	2 mg/kg (n=7)	5 mg/kg (n=3)	9 mg/kg (n=3)	14 mg/kg (n=3)	20 mg/kg (n=6)	Total (N=22)
No. of cycles						
Median (range)	2 (1–22)	2 (1–2)	4 (2–10)	2 (2–3)	3 (2–4)	2 (1–22)
Duration of treatment in days						
Mean (SD)	113.1 (216.7)	29.3 (13.5)	137.0 (115.8)	51.7 (16.8)	71.7 (31.4)	85.3 (127.6)
Median (range)	42 (1–603)	29 (16–43)	99 (45–267)	43 (41–71)	71 (43–102)	43 (1–603)
Reason for discontinuation, n (%)						
AE	1 (14.3)	2 (66.7)	0 (0)	0 (0)	1 (16.7)	4 (18.2)
Progressive disease	4 (57.1)	1 (33.3)	3 (100.0)	3 (100.0)	5 (83.3)	16 (72.7)
Patient withdrawal	1 (14.3)	0 (0)	0 (0)	0 (0)	0 (0)	1 (4.5)

AE, adverse event; SD, standard deviation.

Safety

- Across all doses of AV-203, there was a single DLT that occurred in an 80-year-old patient in the 2-mg/kg cohort
 - Inability to tolerate the study drug (serious grade 3 diarrhea and multiple concurrent AEs)
- The maximum administered dose of 20 mg/kg was determined to be the RP2D
- The most common treatment-emergent and treatment-related AEs (all grade) were diarrhea and decreased appetite (**Table 3**)
- The only grade 3 or greater AEs in >1 patient were anemia, diarrhea, and hypokalemia (2 patients each)

Table 3. All-Grade Treatment-Emergent Adverse Events in ≥20% of Patients and All-Grade Treatment-Related Adverse Events (N=22)

Adverse Event, n (%)	All-Grade	Treatment-Related
Diarrhea	15 (68.2)	13 (59.1)
Decreased appetite	9 (40.9)	7 (31.8)
Hypokalemia	8 (36.4)	4 (18.2)
Dry skin	8 (36.4)	7 (31.8)
Headache	7 (31.8)	0 (0)
Hypomagnesemia	7 (31.8)	6 (27.3)
Dehydration	6 (27.3)	4 (18.2)
Dizziness	6 (27.3)	2 (9.1)
Dyspnea	6 (27.3)	0 (0)
Nausea	6 (27.3)	3 (13.6)
Anxiety	5 (22.7)	1 (4.5)
Depression	5 (22.7)	0 (0)
Fatigue	5 (22.7)	4 (18.2)

PK, ADA, and PD Characteristics

- PK analysis indicates AV-203 levels are approximately dose proportional (**Table 4**)
- No detection of ADAs were observed
- At all AV-203 dose levels, ERBB3 levels rose by approximately 5-fold from baseline by cycle 1 day 15 and sustained these levels through cycle 2 day 15 (**Figure 2**)

Response

- One partial response that lasted 6 cycles was confirmed in Patient 1 (**Figure 3**) in the 9-mg/kg cohort with squamous non-small cell lung cancer (sqNSCLC)
- Stable disease was the best response for 8 patients
 - Patient 2 (**Figure 3**) demonstrated long-term stable disease lasting at least 22 cycles from start of treatment

Figure 2. ERBB3-ECD Serum Levels by (A) Dose Level Cohorts and (B) Fold Change for All Patients Over Baseline.

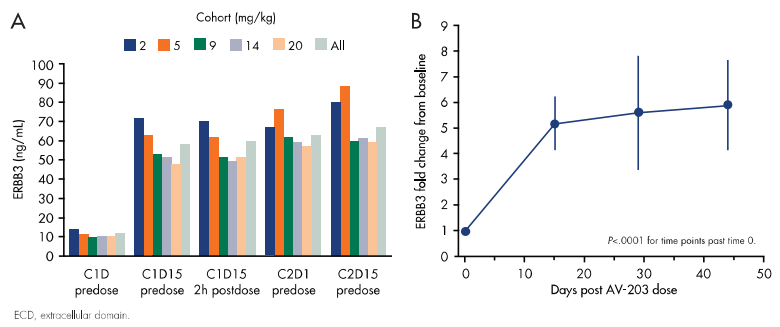
A
Cohort (mg/kg)
2 5 9 14 20 All
ERBB3 (ng/mL)
C1D predose
C1D15 predose
C1D15 2h postdose
C2D1 predose
C2D15 predose
B
ERBB3 fold change from baseline
P<.0001 for time points past time 0.
Days post AV-203 dose

ECD, extracellular domain.

Figure 3. Patient Response Summaries.

Patient 1 – sqNSCLC with partial response (9 mg/kg)

- Diagnosed December 5, 2007
- Prior therapies
 - Cisplatin/etoposide ([neo]adjuvant, May–July 2008)
 - Carboplatin/taxane (June–November 2011)
 - Gemcitabine (March–October 2012)
- Started AV-203 November 2012
 - Partial response detected at the end of cycle 4 and lasted for 6 cycles
 - Reduction of 34% in sum of the longest diameter of target lesion (17% at cycle 2)
- NRG-1 status from baseline tumor specimen: Positive

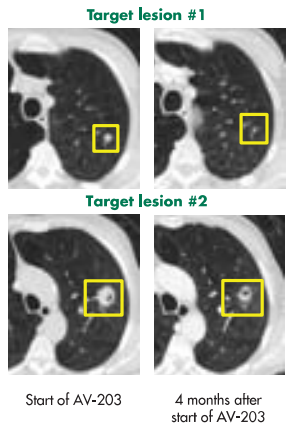
Target lesion #1
Target lesion #2
Start of AV-203
4 months after start of AV-203

Patient 2 – metastatic endometrial biphasic stromal cancer with long-term stable disease (2 mg/kg)

- Diagnosed March 16, 2009
- Prior therapies
 - Combination chemotherapy, including carboplatin and paclitaxel
 - Following progressive disease, treated with investigational anti-endoglin antibody
- Started AV-203 July 2013
 - Received >22 cycles with continued stable disease and no DLTs or dose reductions
 - Non-partial response reduction in target lesions and stable to slightly improved non-target lesions per RECIST
- NRG-1 status from baseline tumor specimen: Negative

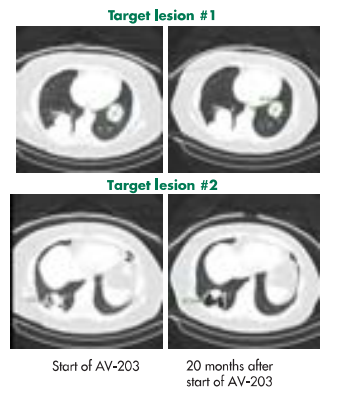
Target lesion #1
Target lesion #2
Start of AV-203
20 months after start of AV-203

DLT, dose-limiting toxicity; NRG, neuregulin; RECIST, Responsive Evaluation Criteria In Solid Tumors; sqNSCLC, squamous non-small cell lung cancer.

Table 4. Pharmacokinetic Characteristics of AV-203

	2 mg/kg (n=7)	5 mg/kg (n=3)	9 mg/kg (n=3)	14 mg/kg (n=3)	20 mg/kg (n=6)
C_{max} (ug/mL)					
Mean (SD)	55.0 (11.8)	110.2 (18.8)	215.3 (15.6)	317.7 (142.3)	555.2 (128.4)
CV%	21.4	17.1	7.2	44.8	23.1
$AUC_{0-\infty}$ (h*ug/mL)					
Mean (SD)	8506.2 (2478.4)	21083.5 (4601.9)	37643.9 (969.1)	50855.6 (17638.7)	93120.4 (15796.3)
CV%	29.1	21.8	23.1	34.7	17.0
CL (mL/h/kg)					
Mean (SD)	0.26 (0.09)	0.25 (0.06)	0.25 (0.07)	0.30 (0.11)	0.22 (0.04)
CV%	34.5	25.0	26.6	36.8	20.1
V_{ss} (mL/kg)					
Mean (SD)	35.4 (5.2)	50.8 (6.2)	61.3 (11.1)	65.4 (20.2)	45.9 (14.5)
CV%	14.8	12.1	18.2	30.8	31.5
Half-life (h)					
Mean (SD)	112.7 (23.7)	153.6 (17.8)	175.5 (35.4)	161.9 (33.8)	148.5 (14.3)
CV%	21.0	11.6	20.2	20.9	9.6

AUC, area under the curve; C_{max}, maximum concentration; CL, clearance; CV, coefficient of variation; SD, standard deviation; V_{ss}, steady state volume of distribution.

NRG-1 Analysis

- Of 20 subjects providing predose samples, 14 were analyzed for NRG-1 by RT-PCR and 12 could be correlated to an evaluable response (**Table 5**)

Table 5. NRG-1 Status in Evaluable Patients

Patient	NRG-1 Status	Tumor Type	Overall Response
1	Positive	sqNSCLC	PR
2	Negative	Stromal tumor-endometrial	Long-term SD
3	Negative	Peritoneal	PD
4	Positive	Squamous cell carcinoma of the skin	PD
5	Negative	sqNSCLC	PD
6	Negative	Rectal adenocarcinoma	SD
7	Indeterminate	Hepatocellular carcinoma	PD
8	Indeterminate	sqNSCLC	PD
9	Negative	SCCHN	PD
10	Negative	Neurendocrine tumor of the stomach	SD
11	Negative	Bladder	SD
12	Negative	Cervical	SD

NRG-1, neuregulin 1; PD, progressive disease; PR, partial response; SCCHN, squamous cell carcinoma of the head and neck; SD, stable disease; sqNSCLC, squamous non-small cell lung cancer.

Conclusions

- Maximum planned dose of AV-203 monotherapy was reached with 1 DLT
 - RP2D is 20 mg/kg intravenously every 2 weeks
 - Most common AEs were diarrhea and decreased appetite
- PK results indicated dose-proportional levels of AV-203
- No detection of ADAs was observed
- PD analysis showed ERBB3 levels were increased approximately 5-fold
- Expression of NRG-1 in the sqNSCLC patient with the partial response may indicate the potential to use NRG-1 as a predictive biomarker for patient selection
- The partial response in a patient with sqNSCLC, as well as the long-term stable disease observed, indicates that AV-203 warrants further investigation as an anticancer therapy

References

1. Aurisicchio L, et al. *Oncotarget*. 2012;3:744–758.
2. Jathal MK, et al. *Immunol Endocr Metab Agents Med Chem*. 2011; 11:131–149.
3. Lee Y, et al. *Acta Biochim Biophys Sin (Shanghai)*. 2014;46:190–198.
4. Vincent S, et al. Presented at: AACR Annual Meeting; March 31–April 4, 2012; Chicago IL. Abstract 2509.
5. Meetze K, et al. Presented at: AACR Annual Meeting; March 31–April 4, 2012; Chicago IL. Abstract 3787.
6. Tyler SM, et al. Presented at: AACR Annual Meeting; April 6–10, 2013; Washington, DC. Abstract 1228.

Acknowledgments

This study was sponsored by AVEO Oncology. Medical writing and editorial assistance was provided by Scientific Connexions and was funded by AVEO Oncology.

PRESENTED AT THE AMERICAN SOCIETY OF CLINICAL ONCOLOGY ANNUAL CONGRESS; MAY 30–JUNE 3, 2014; CHICAGO, IL

Corresponding author: John Sarantopoulos, sarantopoulo@uthscsa.edu. Poster will be available on-line at http://investor.aveooncology.com