AV-203, a Humanized ERBB3 Inhibitory Antibody Inhibits Ligand-Dependent and Ligand-Independent ERBB3 Signaling *in vitro* and *in vivo*

Sylvie Vincent, Christina Fleet, Steven Bottega, Donna McIntosh, William Winston, Ting Chen, Steven Tyler, Kristan Meetze, Solly Weiler, Jeno Gyuris

AVEO Pharmaceuticals, 75 Sidney St, Cambridge, MA, USA

AACR, 2012
Poster # 2509

ERBB3, a member of the EGFR receptor tyrosine kinase (RTK) family, has been implicated in tumor progression and as a path for resistance to standard of care therapies and RTK inhibitor (TKI) drug treatment. Despite its weak kinase activity, ERBB3 is of particular interest due to its ability to heterodimerize with pro-oncogenic RTK partners, such as HER2, EGFR, and MET. Activation of ERBB3 leads to potent activation of the PI3K pathway. We have developed AV-203, a humanized immunoglobulin G1/kappa antibody with potent inhibitory activity against RTK-dependent and ligand-induced activation of ERBB3.

AV-203 specifically binds with high affinity to human ERBB3 and to cynomolgus monkey ERBB3 but not to mouse, allowing for toxicological assessment. AV-203 potently inhibits the binding of the ERBB3 ligand Neuregulin (NRG1/HRG) to ERBB3 and the activation of ERBB3 and its downstream signaling molecule AKT, in response to NRG1 and EGF ligand stimulation. In RTK overexpressing cancer cells, AV-203 decreases steady state activation of ERBB3/PI3K signaling. AV-203 blocks ERBB3/HER2 heterodimer formation in response to NRG1 and can efficiently inhibit the NRG1 dependent proliferation of the human breast cancer cell line, MCF7, and of a BaF3 cell line engineered to express ERBB3 and HER2 (BaF3-ERBB3/HER2). AV-203 is capable of down regulating ERBB3 receptor *in vitro* and *in vivo*. Lastly, AV-203 inhibits tumor growth in a broad spectrum of xenograft models with and without RTK amplifications, such as the pancreatic cancer BXPC3 or HER2 amplified breast cancer MDA-MB-453, respectively.

The first in human trial of AV-203 is planned for 2012.

ERBB3 Pathway Activation

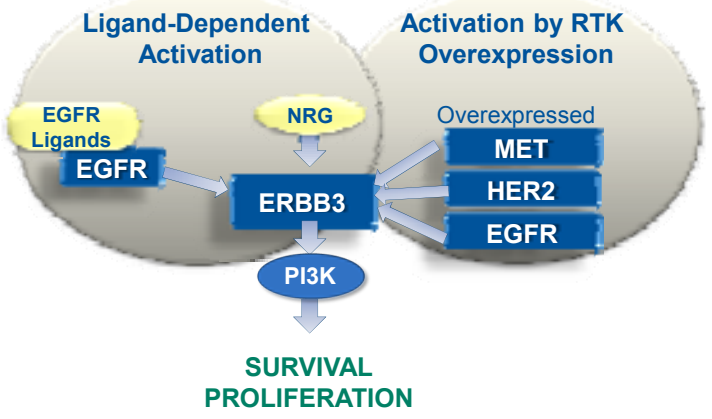
Ligand-Dependent Activation
Activation by RTK Overexpression
EGFR Ligands
EGFR
NRG
ERBB3
Overexpressed
MET
HER2
EGFR
PI3K
SURVIVAL
PROLIFERATION

Kinetic Values of AV-203 Binding to Human ERBB3-ECD

- **AV-203 binds to monomeric human ERBB3 with high affinity (KD=76pM) at 37°C.**
- **A fast on-rate (Ka) and slow off-rate (Kd) were seen at both 25°C and 37°C.**

Temp.	n	Ka Average ($M^{-1}s^{-1}$)	Kd Average (s^{-1})	KD Average (M)
25°C	6	2.6E+06	4.6E-05	**1.8E-11**
37°C	6	3.5E+06	2.7E-04	**7.6E-11**

AV-203 Binding Specificity Determined by ELISA

AV-203 binds to human and cynomolgus ERBB3 but not murine ERBB3

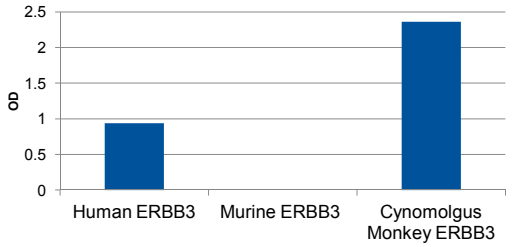
OD
2.5
2
1.5
1
0.5
0
Human ERBB3
Murine ERBB3
Cynomolgus Monkey ERBB3

AV-203 binds specifically to ERBB3

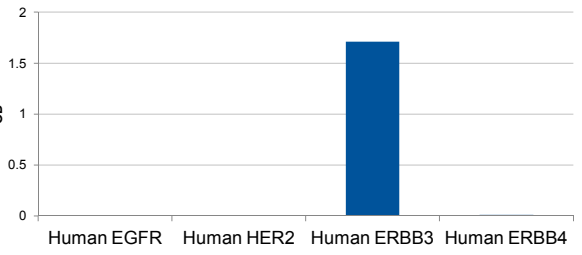
OD
2
1.5
1
0.5
0
Human EGFR
Human HER2
Human ERBB3
Human ERBB4

AV-203 Inhibition of NRG1 Binding to rhERBB3-ECD-Fc

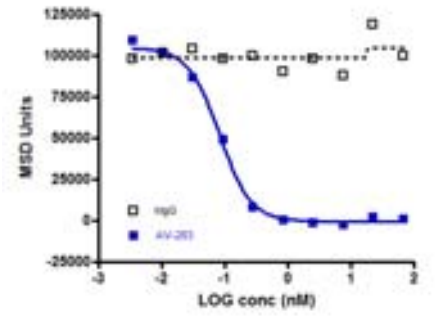
MSD Units
LOG conc (nM)

IC_{50}	n	AVE	STDEV
AV-203	10	0.094	0.023

rhERBB3-ECD-Fc (recombinant human extracellualr domain of ERBB3 with Fc tag)

HER2/ERBB3 Dimer Inhibition

AV-203 can inhibit NRG1-induced ERBB3-HER2 heterodimer formation in MCF7 breast cancer cells

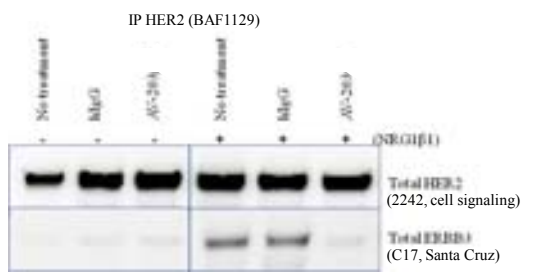
IP HER2 (BAF1129)
(2242, cell signaling)
(C17, Santa Cruz)

Down Regulation of ERBB3

AV-203 down-regulates the ERBB3 receptor over time in HER2 overexpressed MDA-MB-453 breast cancer cells

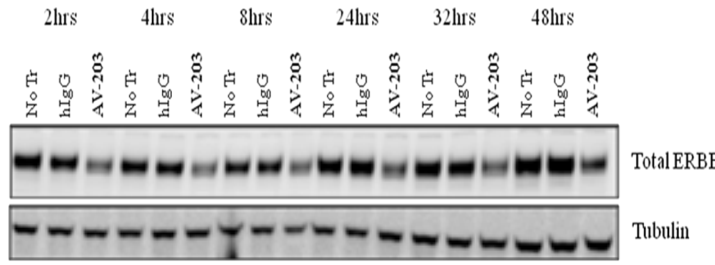
2hrs
4hrs
8hrs
24hrs
32hrs
48hrs
No Tr
hIgG
AV-203
Total ERBB3
Tubulin

AV-203 Inhibition of NRG1 Dependent Proliferation

BaF3 ERBB3/HER2

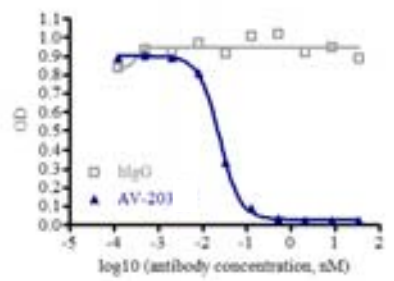
OD
hIgG
AV-203
log10 (antibody concentration, nM)

IC_{50}	n	AVE	STDEV
AV-203	9	0.089	0.072

MCF7

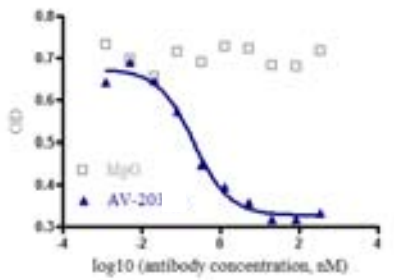
OD
hIgG
AV-203
log10 (antibody concentration, nM)

IC_{50}	n	AVE	STDEV
AV-203	8	0.575	0.583

AV-203 Inhibition of Ligand-Dependent Signaling

AV-203 inhibits EGF- and NRG1β1-dependent ERBB3 signaling in NCI/ADR-res ovarian cancer cells

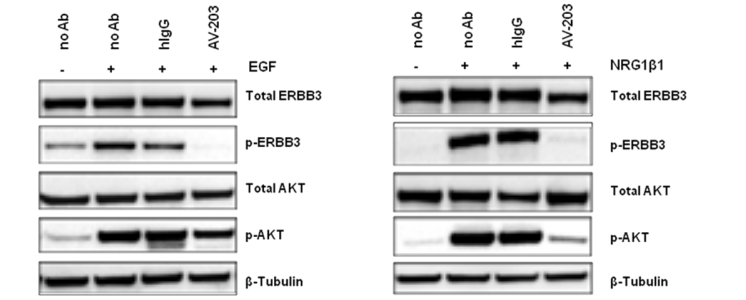
no Ab
no Ab
hIgG
AV-203
EGF
NRG1β1
Total ERBB3
p-ERBB3
Total AKT
p-AKT
β-Tubulin

AV-203 Inhibition of RTK Dependent Signaling

AV-203 inhibits phosphorylation of ERBB3 and AKT in exponentially growing HER2-positive MDA-MB-453 breast cancer cells

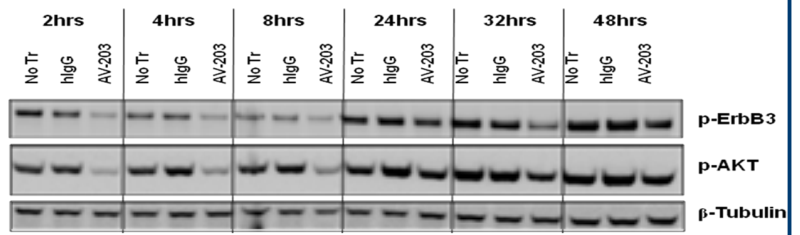
2hrs
4hrs
8hrs
24hrs
32hrs
48hrs
No Tr
hIgG
AV-203
p-ErbB3
p-AKT
β-Tubulin

AV-203 Anti-Tumor Activity in RTK Amplified and Non-Amplified Tumors

AV-203 inhibits tumor growth of BXPC3 xenografts

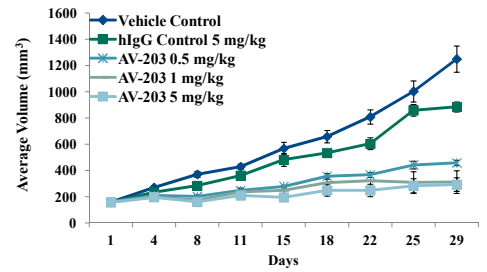
Vehicle Control
hIgG Control 5 mg/kg
AV-203 0.5 mg/kg
AV-203 1 mg/kg
AV-203 5 mg/kg
Average Volume (mm³)
Days

AV-203 inhibits tumor growth of HER2 overexpressing MDA-MB-453 xenografts

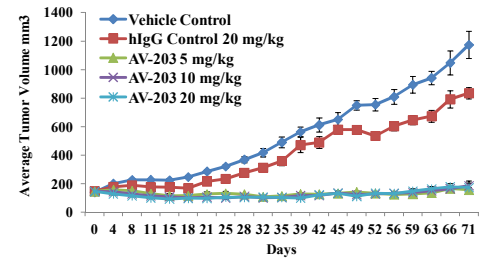
Vehicle Control
hIgG Control 20 mg/kg
AV-203 5 mg/kg
AV-203 10 mg/kg
AV-203 20 mg/kg
Average Tumor Volume mm3
Days

AV-203 also exhibits significant tumor growth inhibition of breast, lung, kidney, head and neck, pancreatic, and ovarian xenografts

Summary

AV-203 is a potent humanized ERBB3 inhibitory antibody that neutralizes NRG1 binding and promotes the downregulation of the ERBB3 pathway

AV-203 inhibits RTK overexpression-dependent and ligand-induced activation of ERBB3

AV-203 inhibits cell proliferation in response to NRG1

AV-203 demonstrates tumor growth inhibition in tumor models with different modes of ERBB3 activation

AACR 103rd Annual Meeting, 2012, Chicago Illinois