Pharmacokinetics and Pharmacodynamics of AV-203, a Humanized anti-ERBB3 Antibody

AVEO PHARMACEUTICALS, INC.

Kristan Meetze, Steven Tyler, Christina Fleet, James Gifford, Sylvie Vincent, Jeno Gyuris

AVEO Pharmaceuticals, 75 Sidney St, Cambridge, MA, USA

AACR, 2012
Poster # 3787

Abstract:

ERBB3 is widely expressed in human carcinomas, and its overexpression is associated with poor prognosis in patients with various carcinomas, (i.e., breast, ovarian, prostate, colorectal, pancreatic, gastric, and head and neck cancers). The presence of ERBB3 correlates with local to distal metastasis in lung, gastric, and colorectal cancers as well as bone invasion in prostate cancer. Activation of ERBB3 is also implicated in the development of resistance to current cancer treatments. Due to its lack of kinase activity, the activation of the ERBB3 receptor is dependent on heterodimerization with active receptor tyrosine kinases (RTKs). The recruitment of ERBB3 into active, heterodimer complexes is mediated by its ligand Neuregulin-1 (NRG-1) or by amplified, over expressed RTKs in a ligand independent manner. Therefore, ERBB3 can crosstalk with most major receptors involved in cancer development and maintenance such as epidermal growth factor receptor (EGFR), human epidermal growth factor receptor 2 (HER2), and c-MET.

AV-203 is a potent, humanized anti-ERBB3 antibody that inhibits both ligand-dependent and independent activation of ERBB3 both *in vitro* and *in vivo*. Pharmacokinetics (PK) and pharmacodynamics (PD) of AV-203 were characterized in mice using the A549 non-small cell lung cancer xenograft model. AV-203 administered IV in mice exhibits acceptable pharmacokinetics supporting preclinical efficacy studies. AV-203 administered IV in A549 NSCLC xenograft tumor bearing mice had lower serum AUC than naïve mice, demonstrating that the presence of human ERBB3 may alter the PK parameters of AV-203. In evaluating pharmacodynamics *in vivo*, AV-203 was able to down regulate total ERBB3 and ERBB3 signaling in A549 tumors in a time-dependent manner. Inhibition of ERBB3 signaling correlated with significant dose-dependent tumor growth inhibition in this model. Dose scheduling studies with the constant AV-203 dose of 2.5 mg/kg revealed that the most efficacious schedule is the more frequent dosing at Q3D. In comparing the total dose of 10 mg/kg per 14 day cycle, at varying dose per injection and frequency, AV-203 resulted in significant tumor growth inhibition at all dose schedules. These data conclude that the efficacy of AV-203 is driven by total drug exposure and that AV-203 is not dependent on C_{max} for its anti-tumor activity.

AV-203 Pharmacokinetics in Naïve and A549 Xenograft Tumor-Bearing NCR Nude Mice

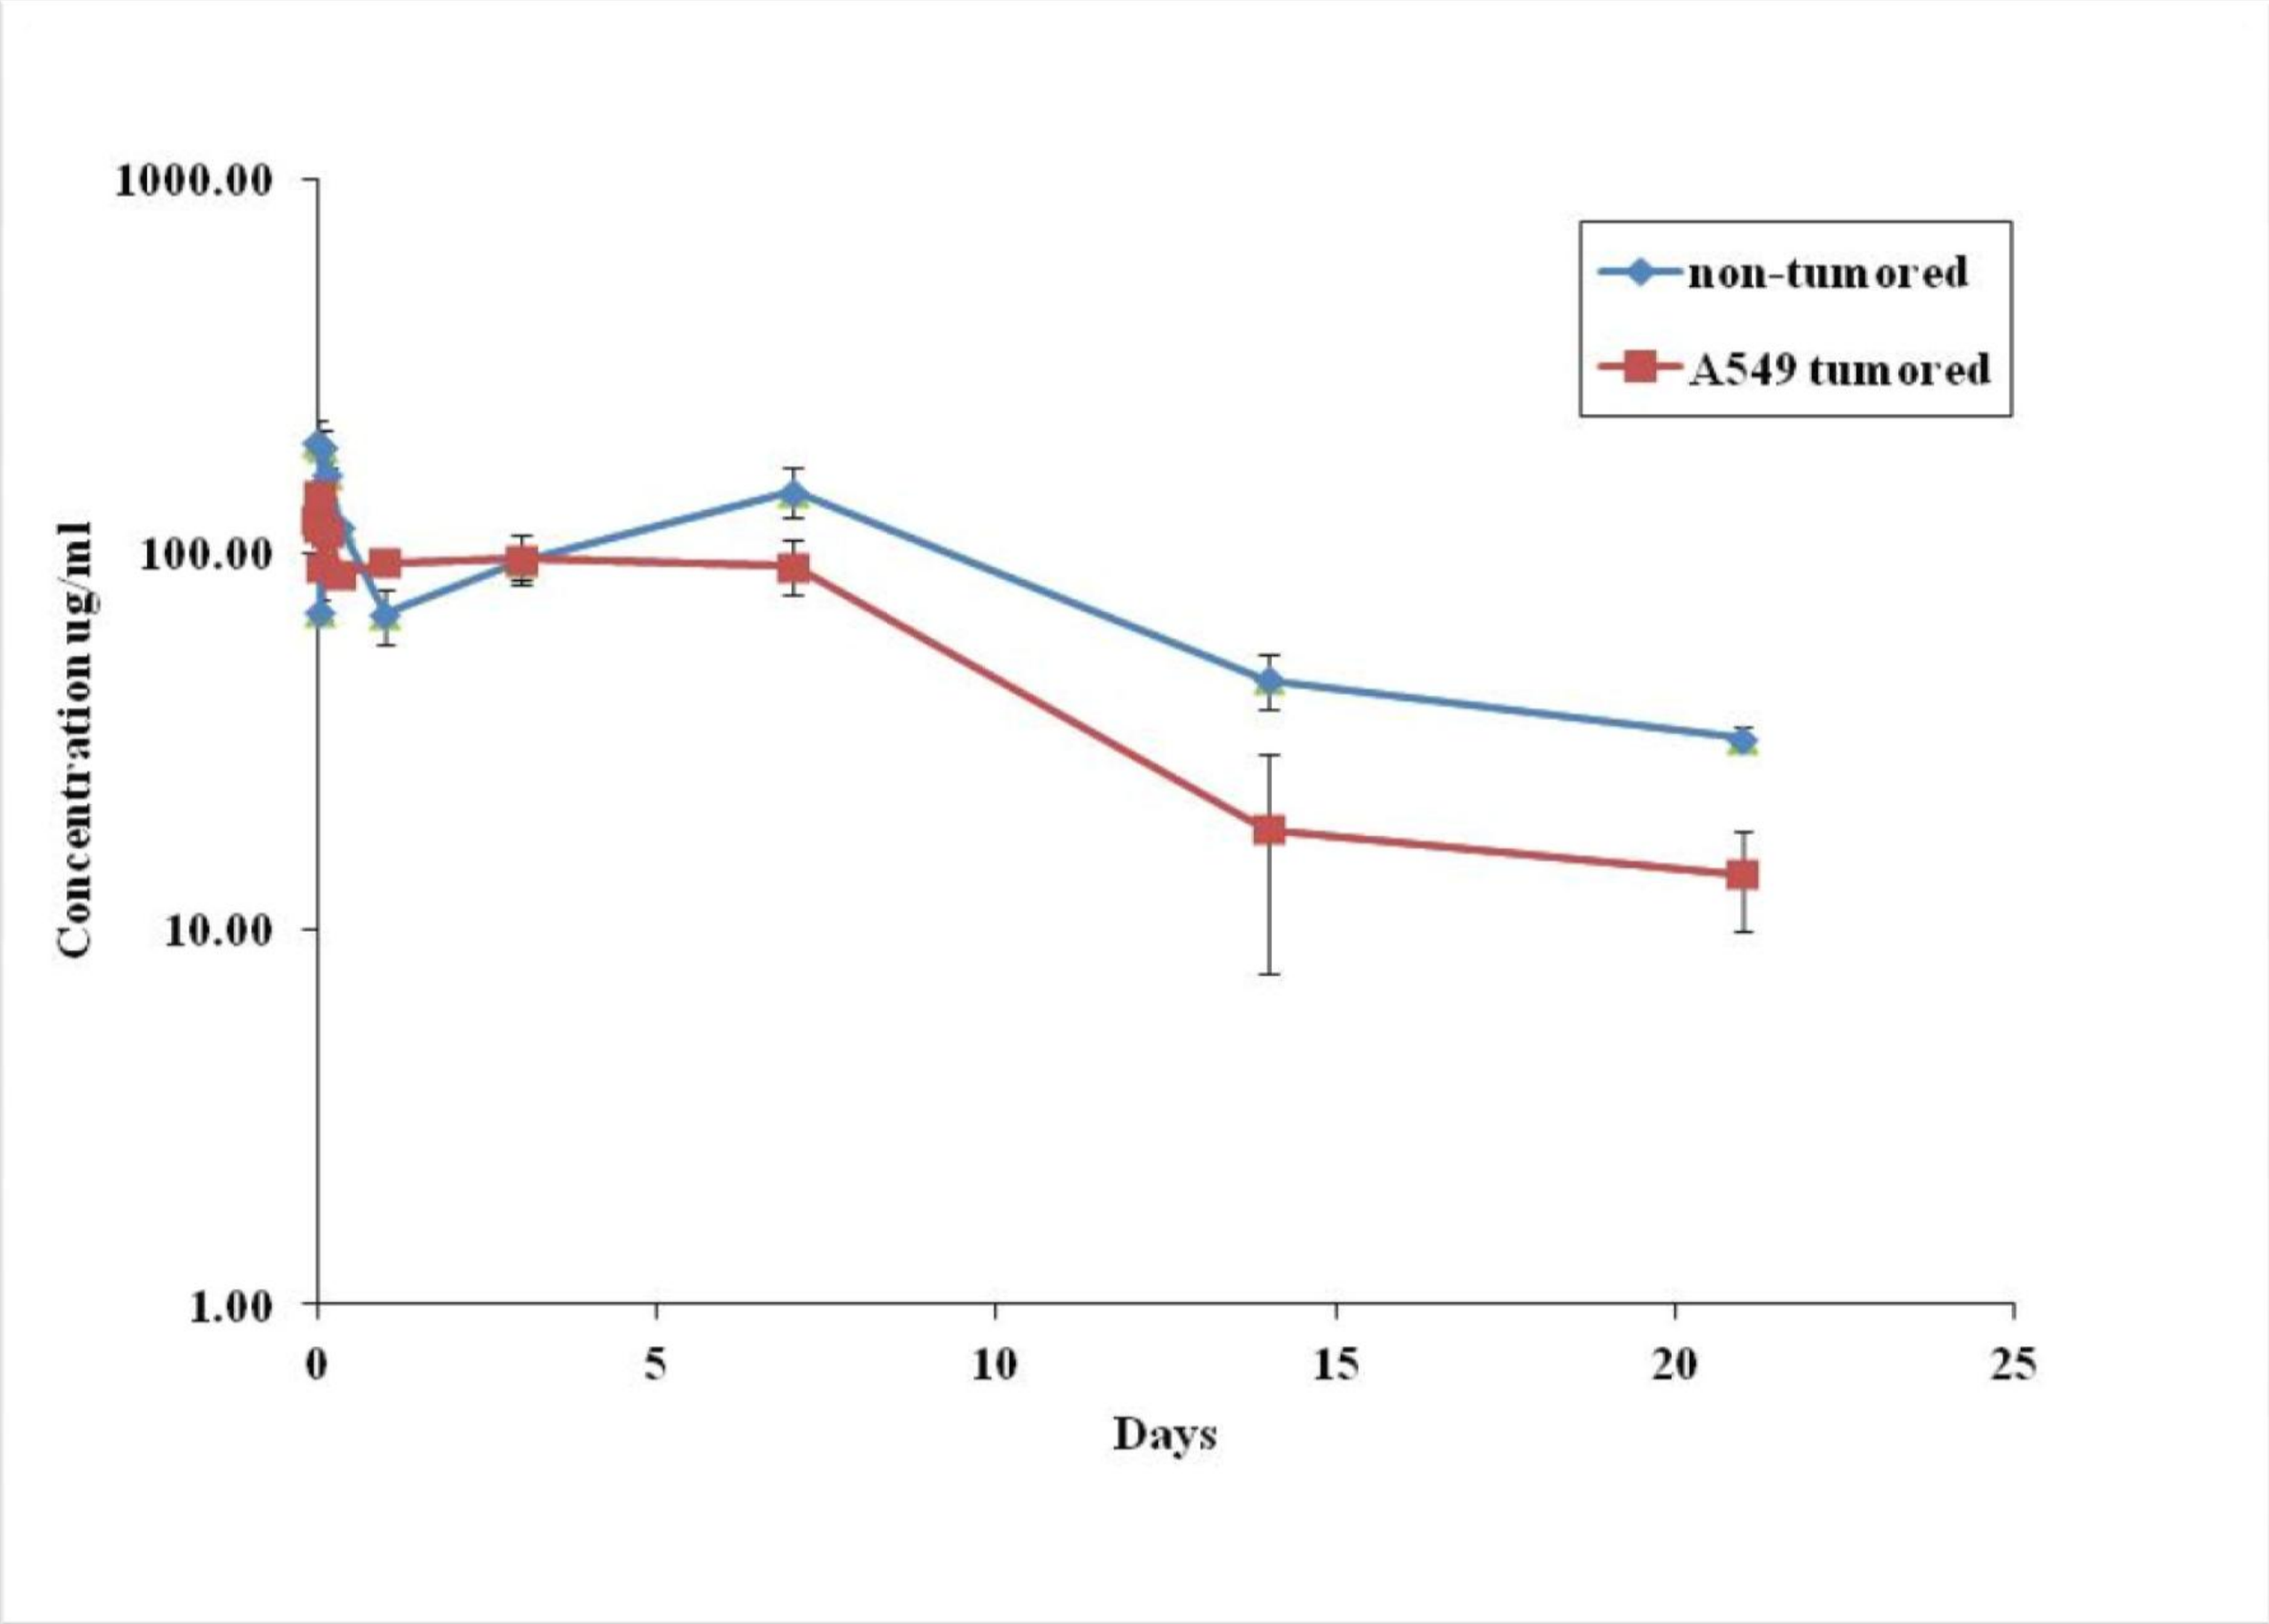
1000.00
100.00
10.00
1.00
Concentration ug/ml
0
5
10
15
20
25
Days
non-tumored
A549 tumored

Pharmacokinetic Parameters of AV-203 After a Single 10 mg/kg Dose in Naïve and A549 Xenograft Tumor-Bearing NCR Nude Mice

Group	t1/2, days	Cmax, µg/mL	AUC, day*µg/mL	Vd, mL/kg	Cl, mL/day/kg
Naïve	12.5	200.0	2292.0	78.9	4.4
A549 Tumor Bearing	6.6	142.8	1305.3	73.5	7.7

Tumor Growth Inhibition of A549 Xenografts, at the Same AV-203 Dose per Injection with Varying Schedules

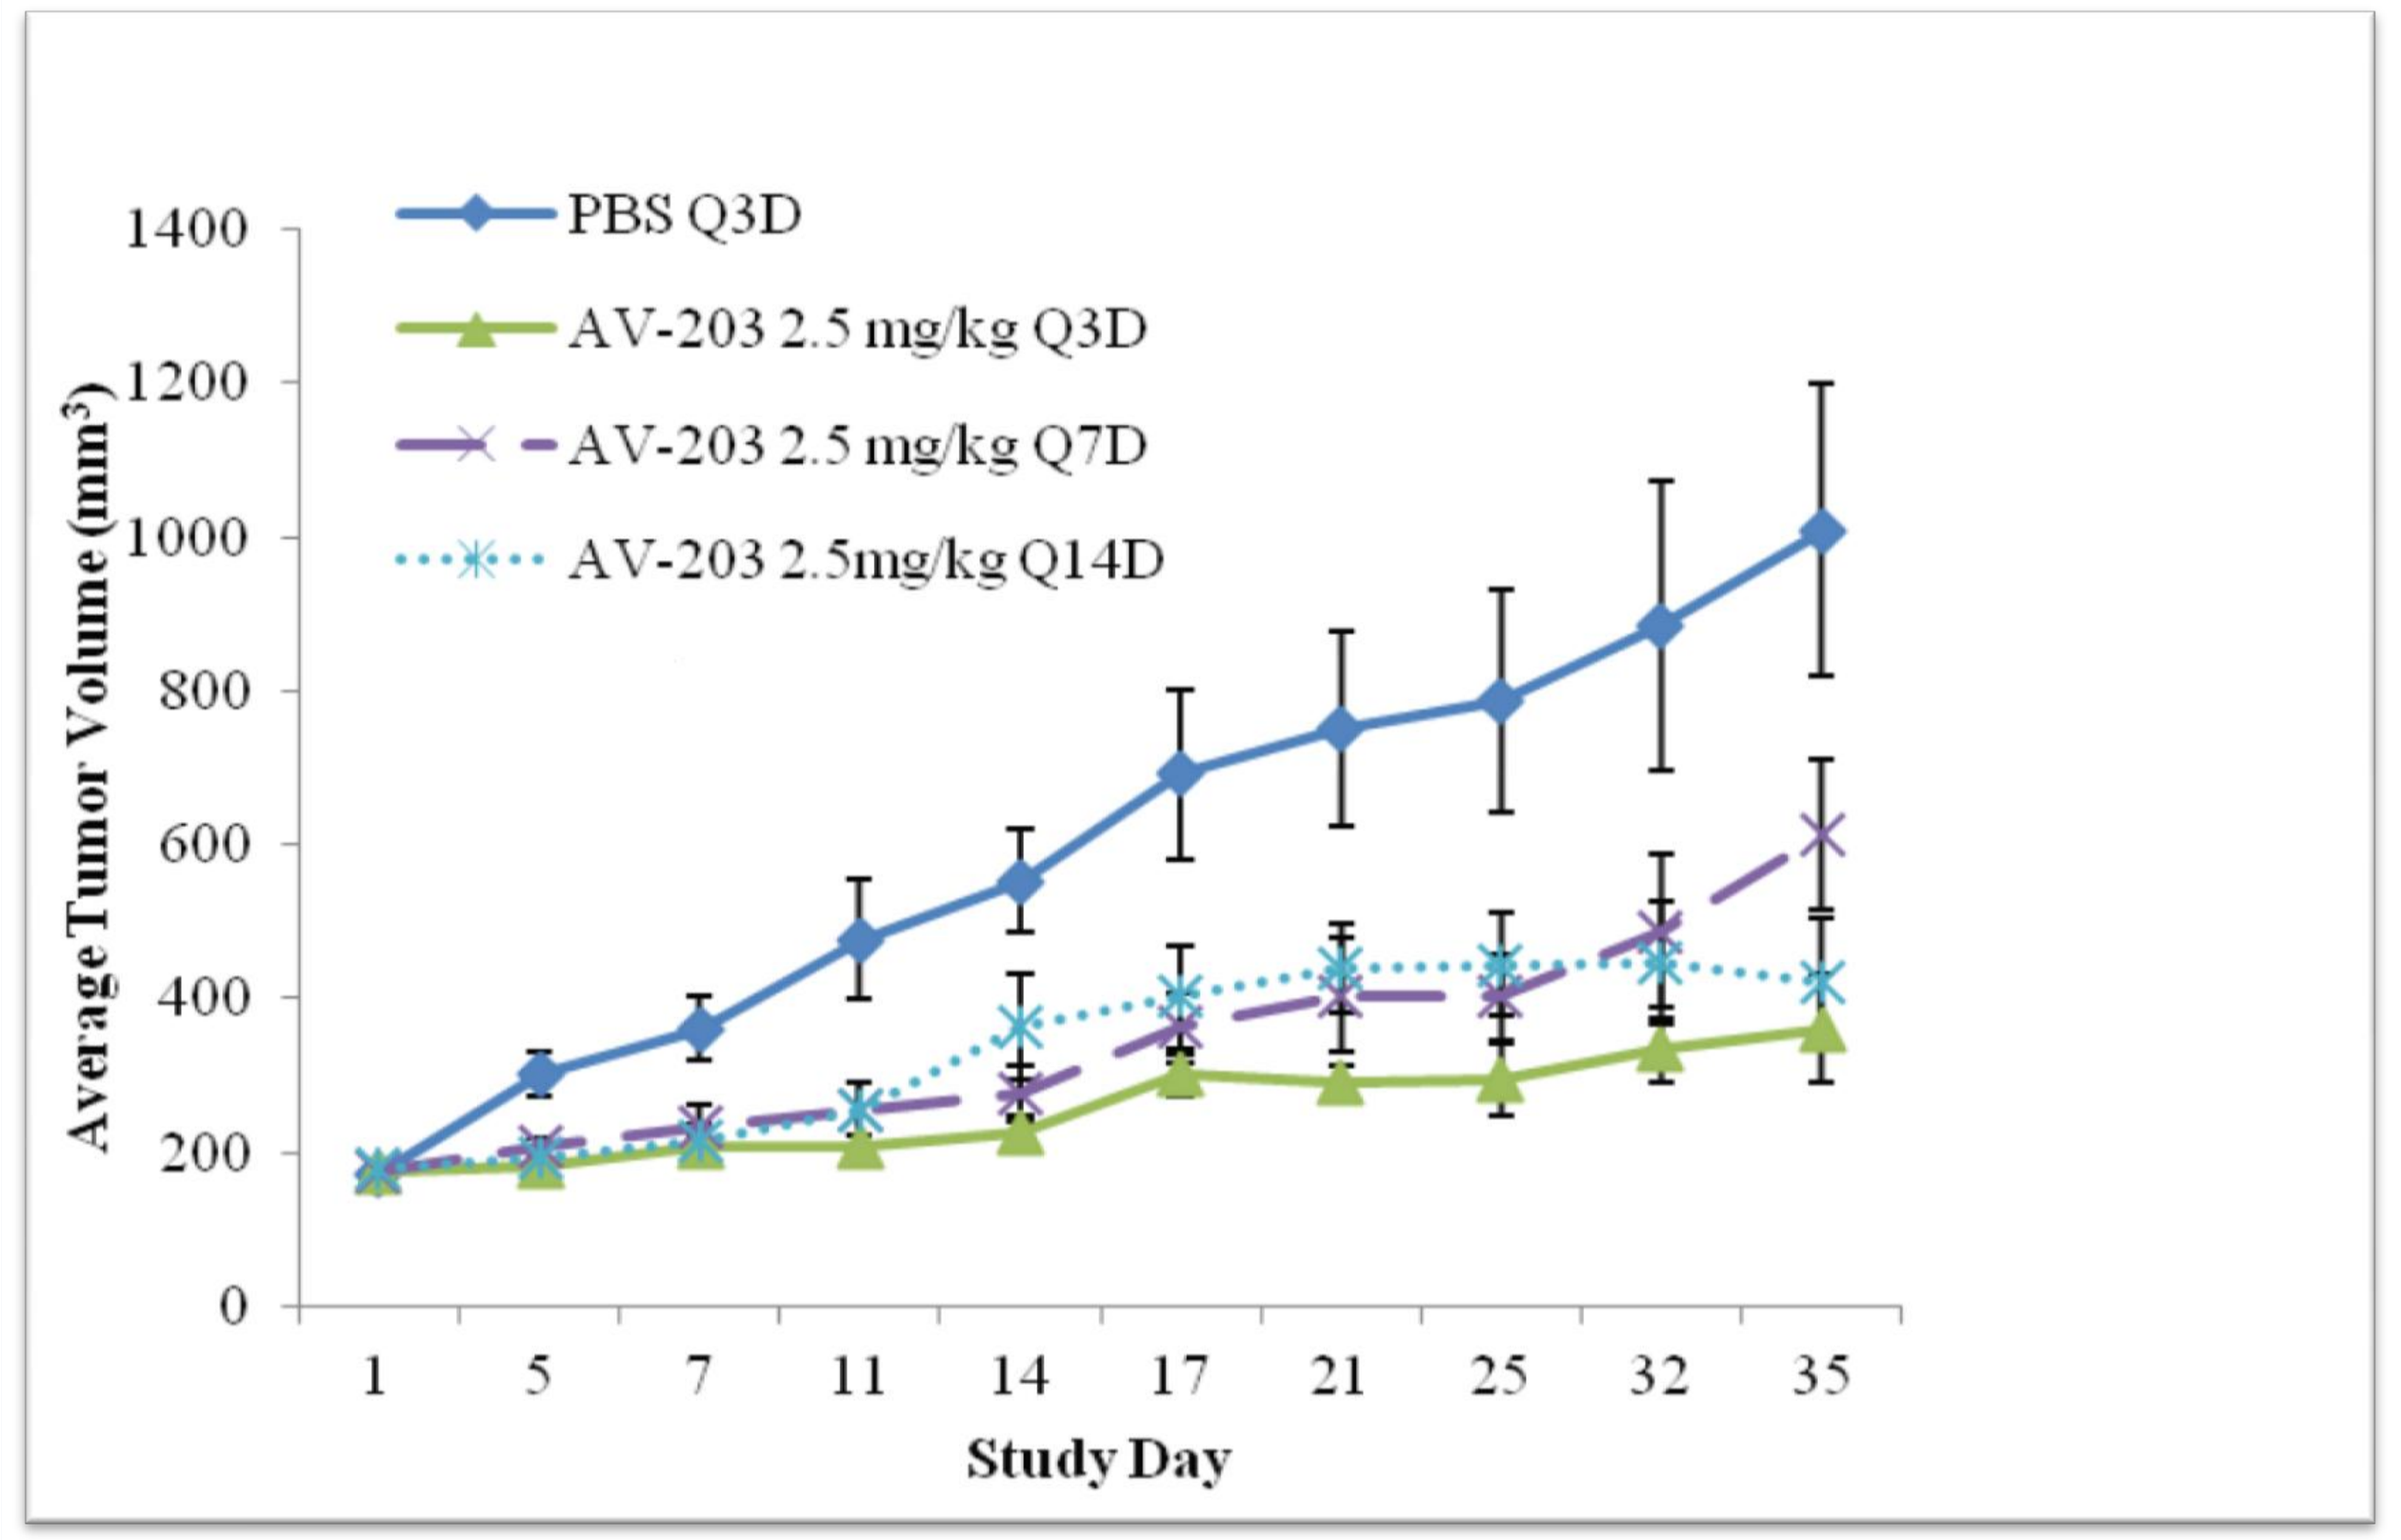
PBS Q3D
AV-203 2.5 mg/kg Q3D
AV-203 2.5 mg/kg Q7D
AV-203 2.5mg/kg Q14D
Average Tumor Volume (mm³)
0
200
400
600
800
1000
1200
1400
1
5
7
11
14
17
21
25
32
35
Study Day

AV-203 when dosed at 2.5 mg/kg on varying schedules resulted in the most efficacious schedule being the most frequent dosing at Q3D.

Tumor Growth Inhibition of A549 Xenografts at the Same AV-203 Dose per 14-day Cycle with Varying Schedules

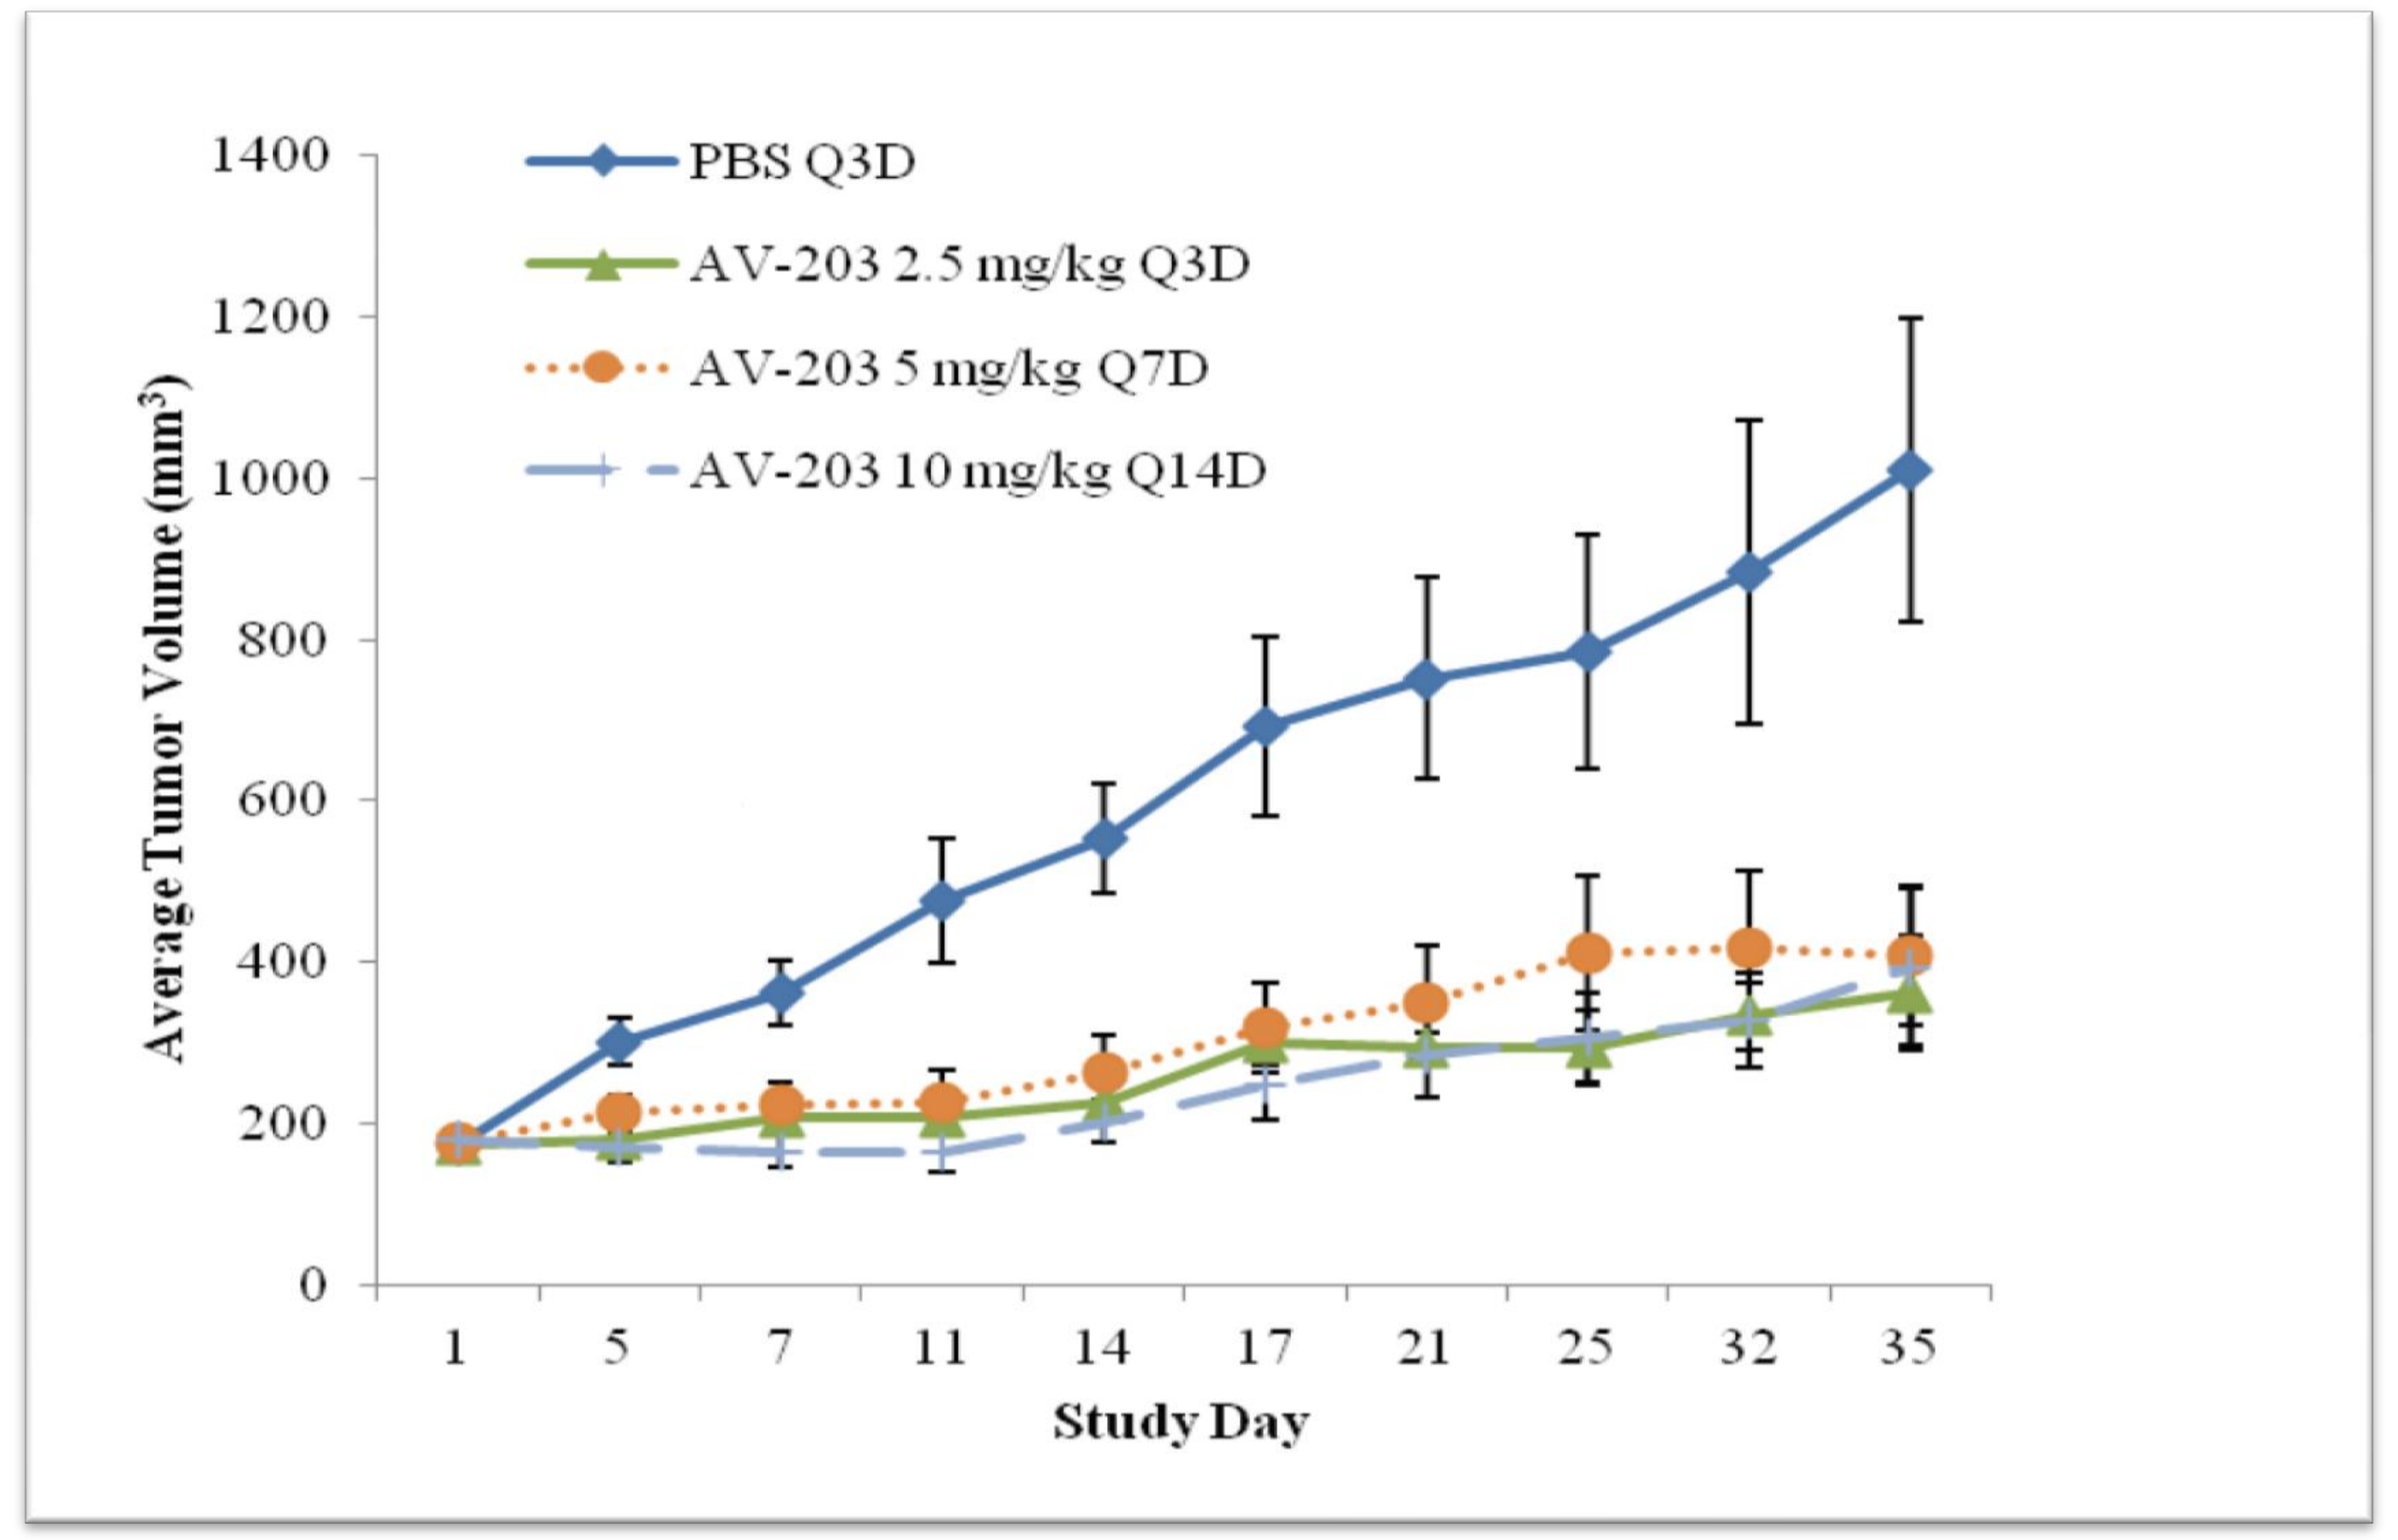
PBS Q3D
AV-203 2.5 mg/kg Q3D
AV-203 5 mg/kg Q7D
AV-203 10 mg/kg Q14D
Average Tumor Volume (mm³)
0
200
400
600
800
1000
1200
1400
1
5
7
11
14
17
21
25
32
35
Study Day

In comparing the total dose of 10 mg/kg per 14-day cycle, at varying dose per injection and varying frequency, AV-203 resulted in similar tumor growth inhibition at all dose schedules.

Summary

AV-203 exhibited acceptable pharmacokinetics in mice supporting preclinical efficacy studies dosing at twice weekly.

Tumor-bearing mice had lower serum AUC, shorter t1/2, and faster CL than naïve mice, suggesting the presence of the target human ERBB3 may increase the CL of AV-203.

AV-203 at 2.5 mg/kg, at varying frequency resulted in the most efficacious being the most frequently dosed at Q3D.

AV-203 at 10 mg/kg, total dose, per 14-day cycle, at varying dose per injection and varying frequency resulted in similar tumor growth inhibition at all dose schedules.

Combined, this data suggested that anti-tumor activity of AV-203 depends on total drug exposure and not on Cmax.

AV-203 down regulates total ERBB3 and pERBB3 signaling in A549 tumors in a time-dependent manner, after a single 20 mg/kg dose.

AV-203 down regulates pAKT signaling in A549 tumors in a time-dependent manner, after a single 20 mg/kg dose.

Mean Total ERBB3 After a Single Dose in A549 Xenograft Tumor-Bearing NCR Nude Mice

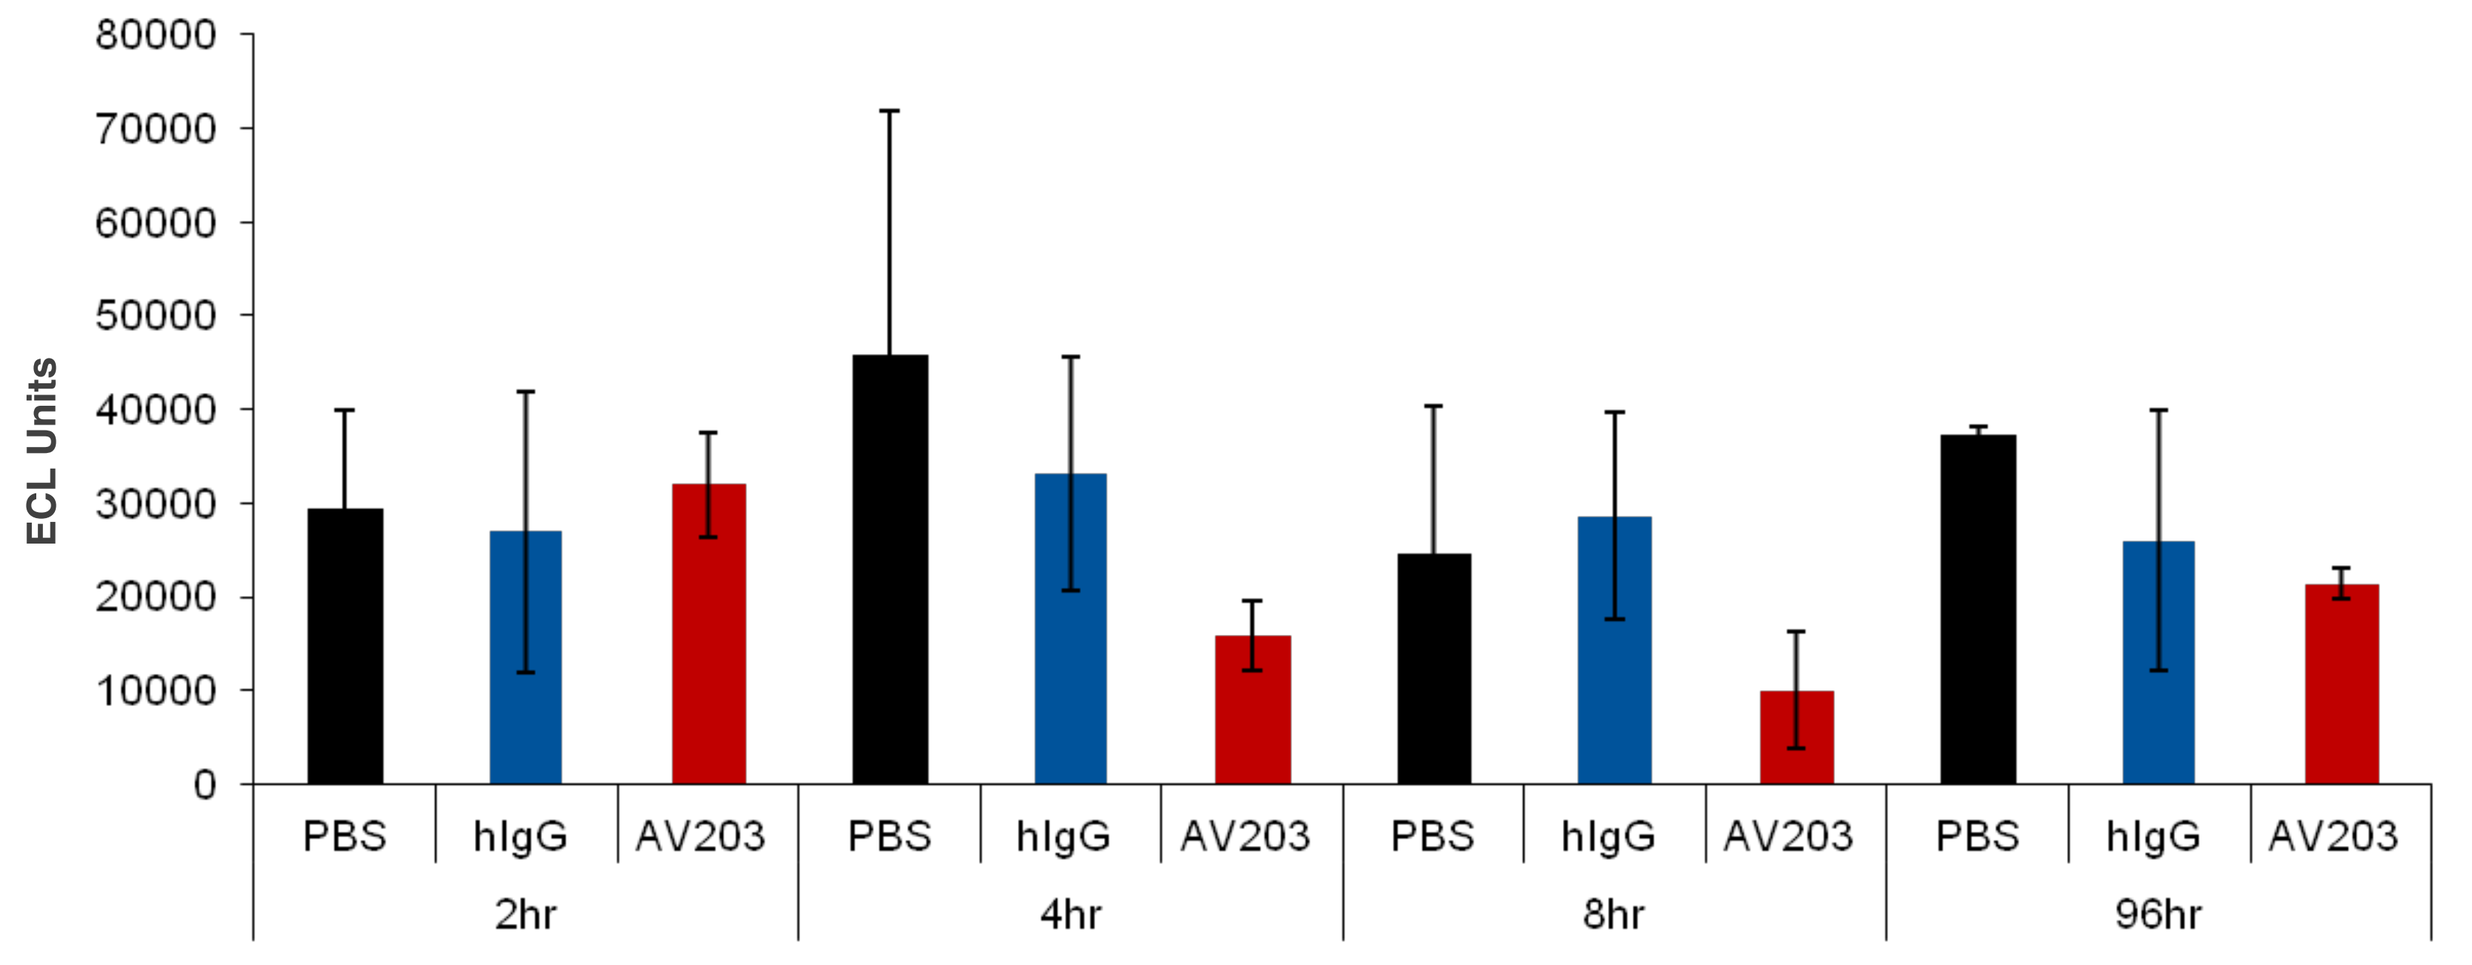
ECL Units
0
10000
20000
30000
40000
50000
60000
70000
80000
PBS
hIgG
AV203
2hr
4hr
8hr
96hr

AV-203 down regulates tERBB3 signaling after a single 20 mg/kg dose from 4 to 96 hours in MSD assays.

Mean pERBB3 After a Single Dose in A549 Xenograft Tumor-Bearing NCR Nude Mice

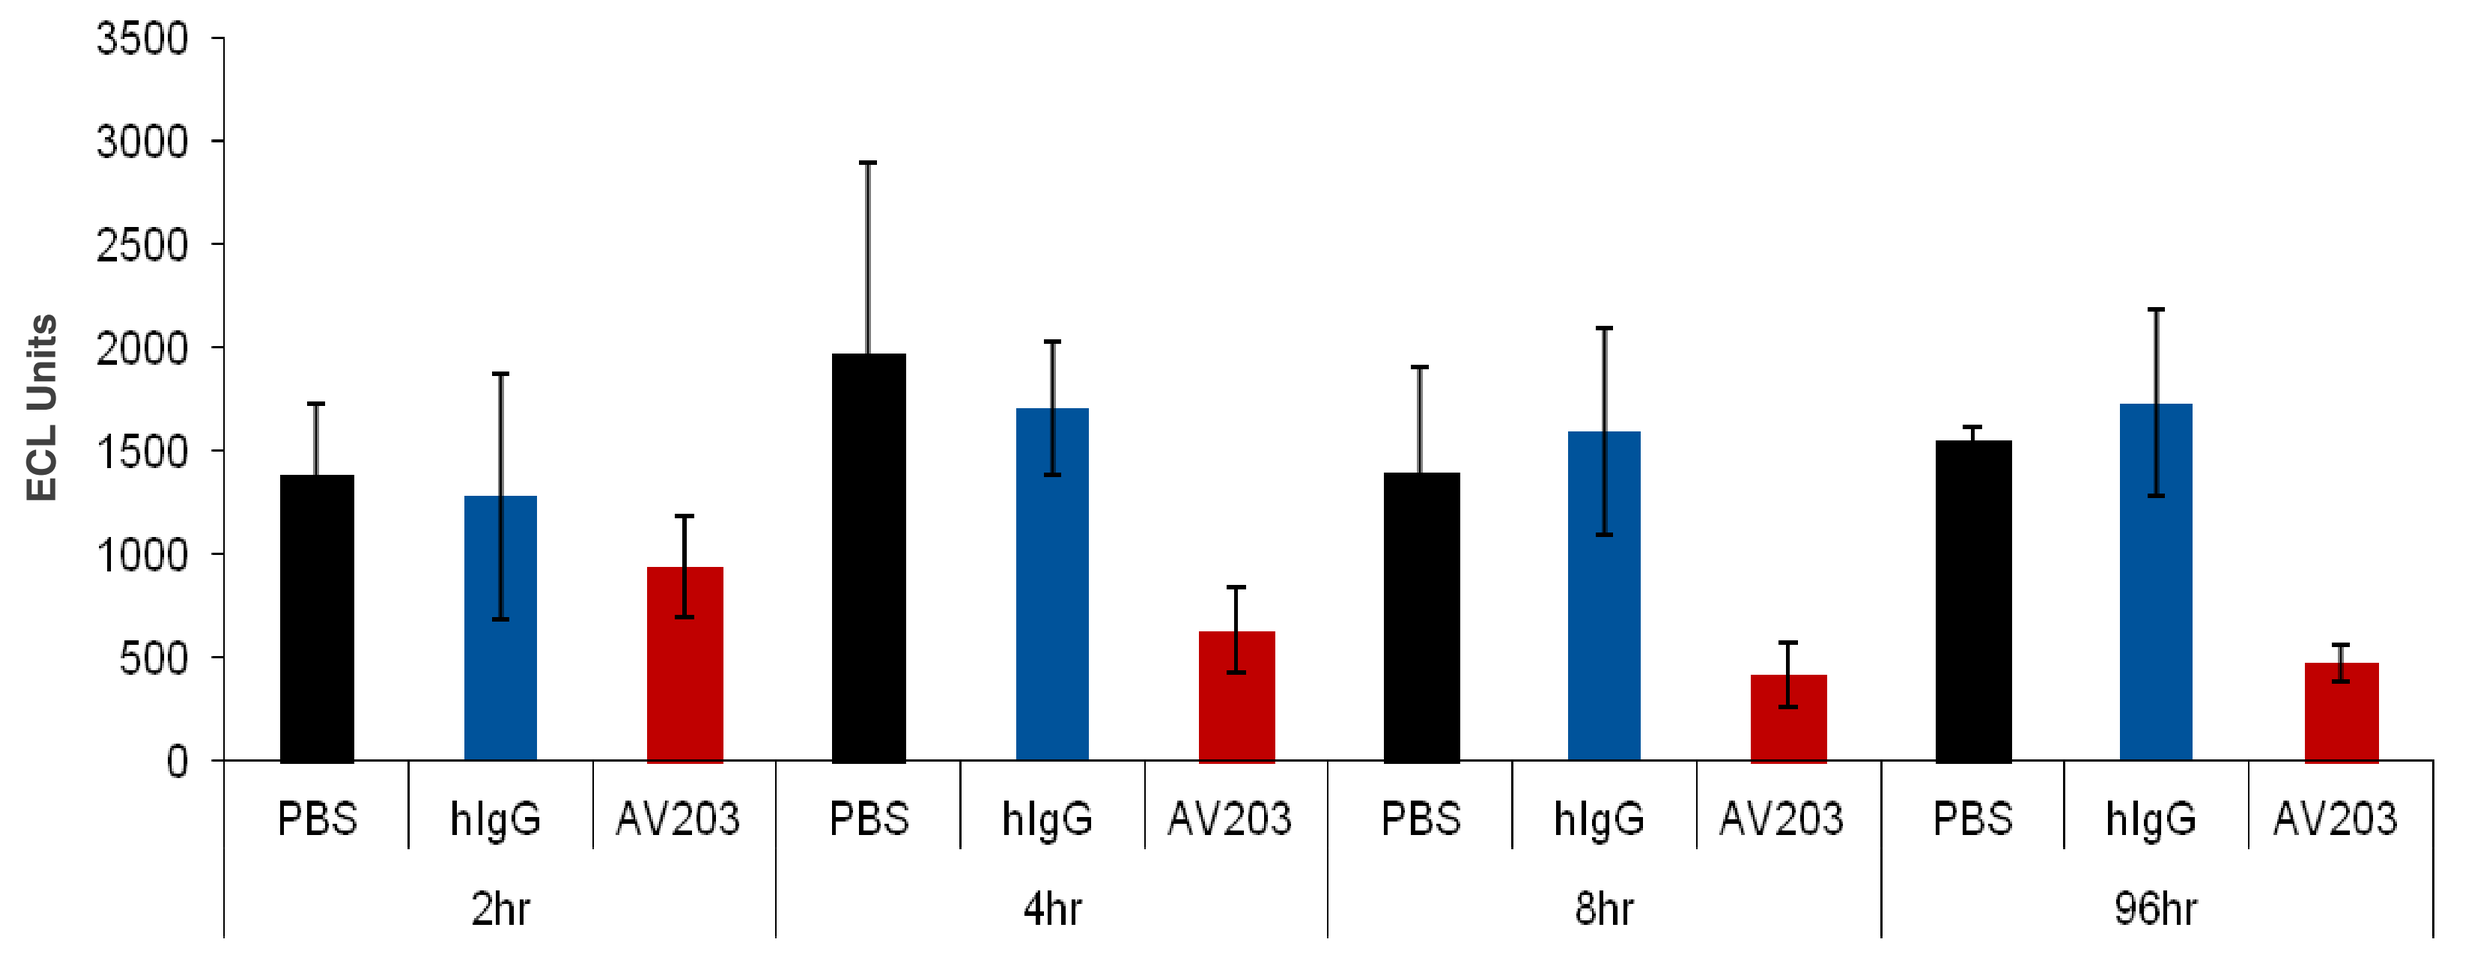
ECL Units
0
500
1000
1500
2000
2500
3000
3500
PBS
hIgG
AV203
2hr
4hr
8hr
96hr

AV-203 down regulates pERBB3 signaling after a single 20 mg/kg dose from 4 to 96 hours in MSD assays.

Mean pAKT After a Single Dose in A549 Xenograft Tumor-Bearing NCR Nude Mice

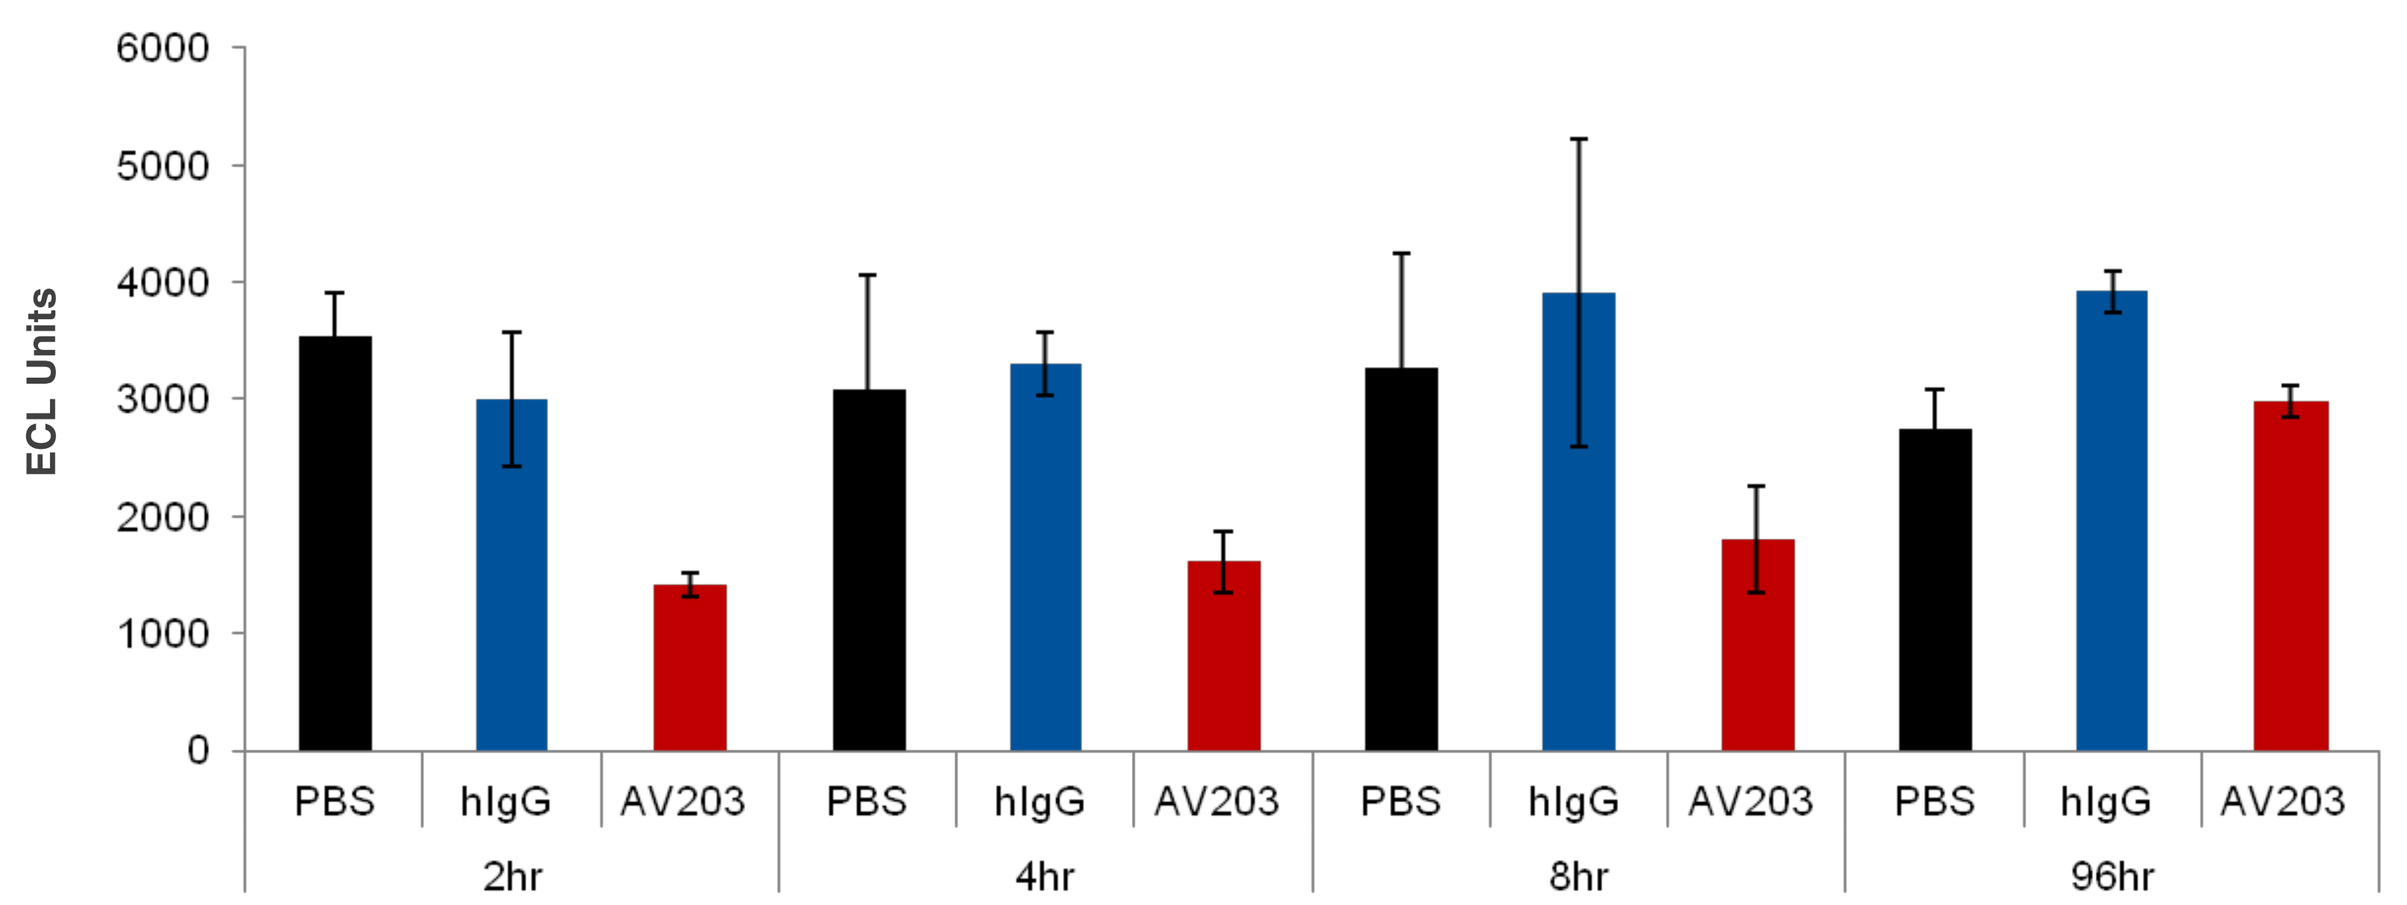
ECL Units
0
1000
2000
3000
4000
5000
6000
PBS
hIgG
AV203
2hr
4hr
8hr
96hr

AV-203 down regulates pAKT signaling after a single 20 mg/kg dose from 2 to 8 hours, with return to normal by 96 hours in MSD assays.

AACR 103rd Annual Meeting, 2012, Chicago Illinois